Biomarkers for Patient Stratification and Target Engagement in ALS Patients with TDP-43 Pathology

Sandy Hinckley[1], Hafiz Mohmmadabdul[1], Daniel Elbaum[1]
[1]QurAlis, Cambridge Massachusetts, United States

Abstract

Patient identification and stratification as well as early identification of target engagement can be crucial to the development of effective therapies in ALS. Multiple prior studies have shown that Stathmin-2 (STMN2) is depleted in the spinal cords of ALS patients. However, it was only recently discovered that TDP-43 mislocalization leads to alternative splicing of a number of genes including Stathmin-2. Stathmin-2, previously named SCG10, is known to play an important role in axonal growth and maintenance, and restoration of Stathmin-2 expression in neurons with TDP-43 nuclear depletion is capable of rescuing axonal health. Intriguingly, in the case of Stathmin-2, the splicing changes seen in patients and human cell models are not recapitulated in lower species including rodents and non-human primates. Given this dichotomy, there is no animal model that is appropriate to assist in human dose prediction. Thus, a clinical dose will be based on efficacy in cell-based assays. With this paradigm it is crucial to be able to evaluate target engagement rapidly in clinical studies. We have developed Stathmin-2 based biomarker assays in human biofluids and characterized these markers in ALS patients and healthy individuals. These assays could be useful to identify patients with TDP-43 proteinopathy and future targeting of these patients for precision medicine-based therapeutics.

Background

TAR DNA binding protein-43 (TDP-43) is a nuclear RNA/DNA binding protein involved in RNA regulation and metabolism. Approximately 97% of amyotrophic lateral sclerosis (ALS) cases have nuclear clearing or insoluble TDP-43 cytoplasmic aggregates present in the brain and/or spinal cord, making this proteinopathy an attractive target. The loss of nuclear TDP-43 results in the mis-splicing of several genes, including *Stathmin-2*. Stathmin-2 is a tubulin stabilizing/destabilizing protein that is required for axonal stability, Golgi function, mitochondrial motility and neuromuscular junction innervation.

In order to identify ALS patients with TDP-43 pathology and support STMN2 target engagement, QurAlis is building a Biomarker platform to identify levels of STMN2 RNA and protein in human biofluids.

Therapeutic Hypothesis

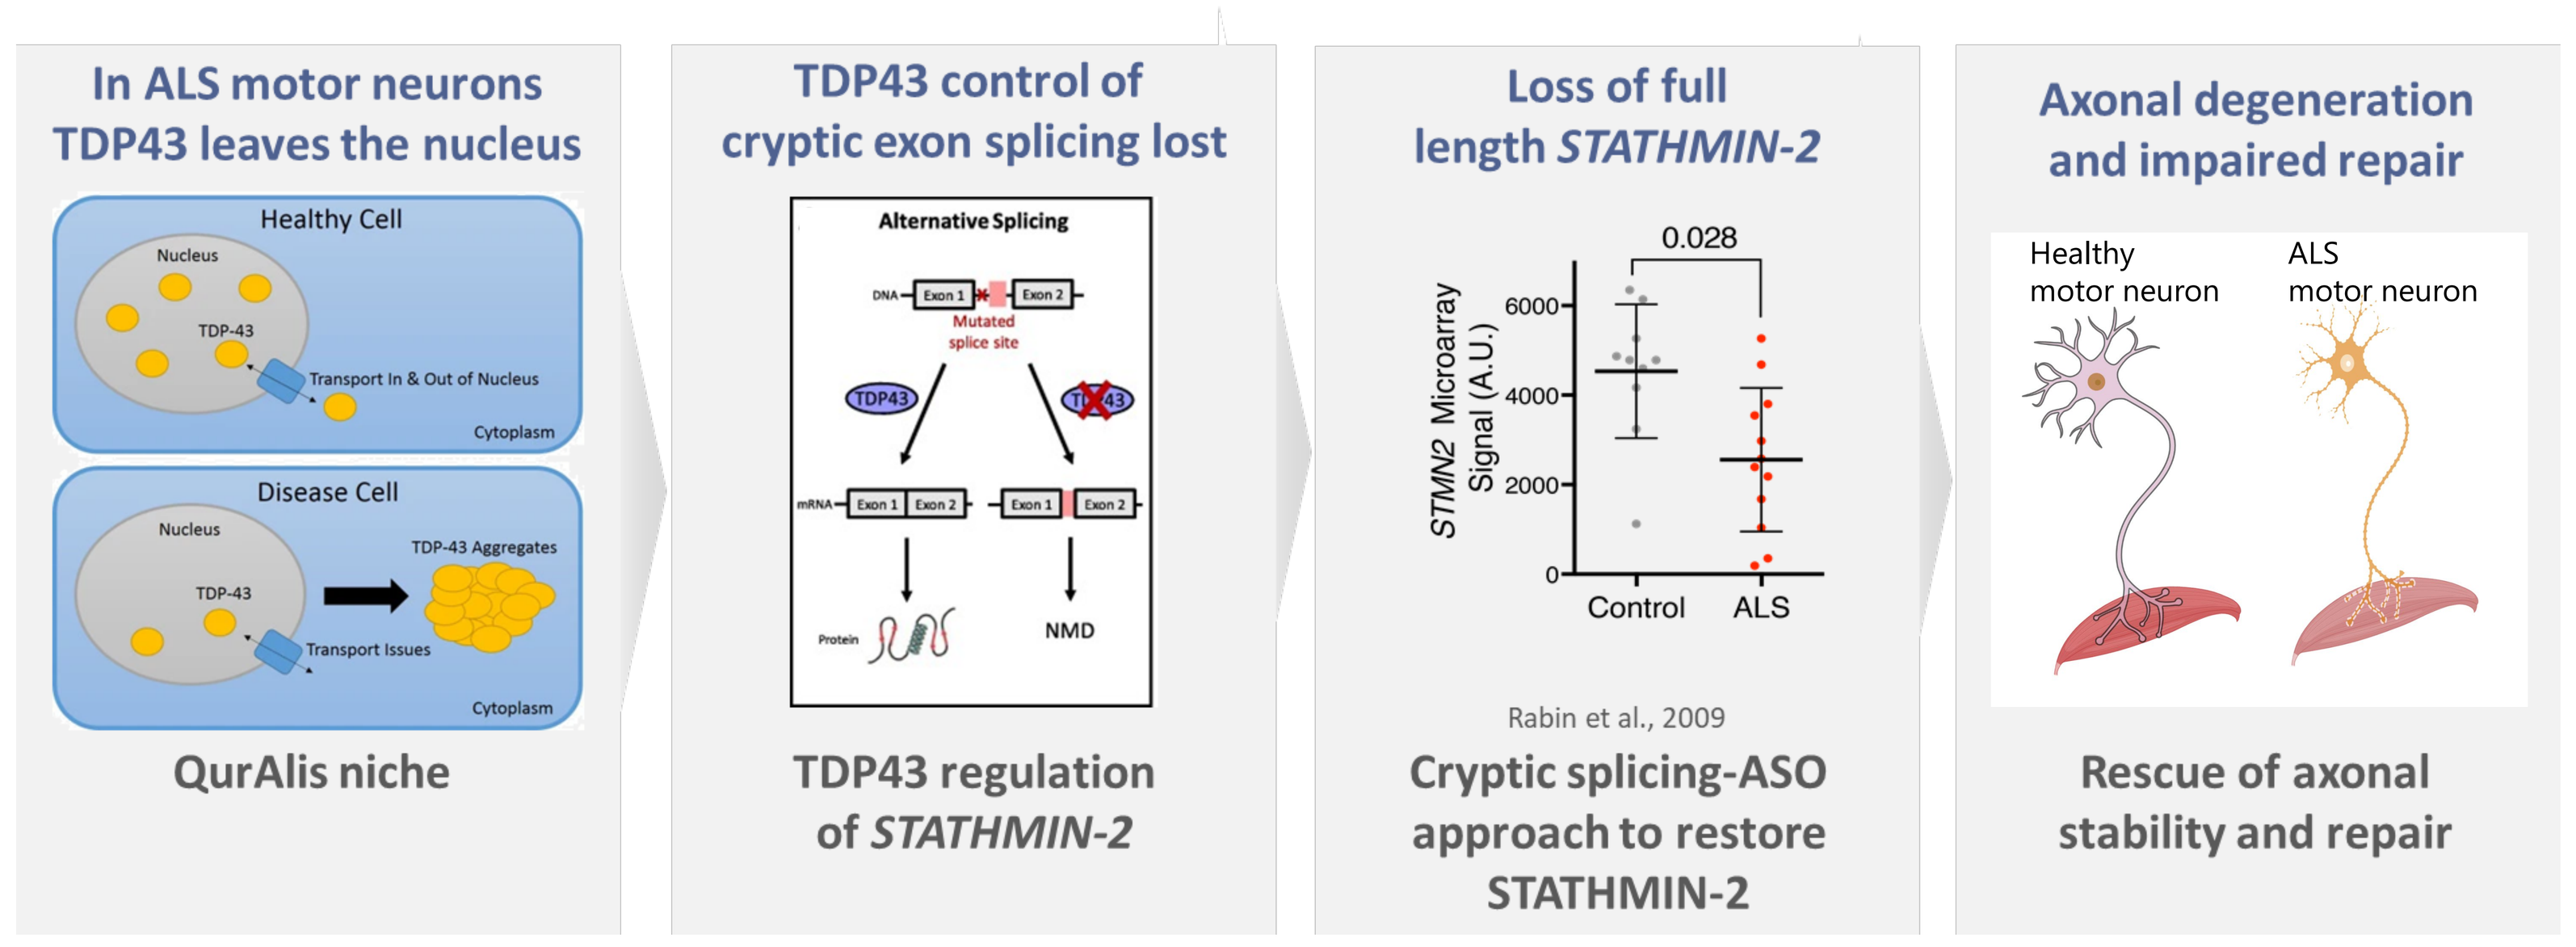
In ALS motor neurons TDP43 leaves the nucleus
Healthy Cell
Nucleus
TDP-43
Transport In & Out of Nucleus
Cytoplasm
Disease Cell
TDP-43 Aggregates
Transport Issues
QurAlis niche
TDP43 control of cryptic exon splicing lost
Alternative Splicing
Mutated splice site
NMD
TDP43 regulation of STATHMIN-2
Loss of full length STATHMIN-2
0.028
STMN2 Microarray Signal (A.U.)
Control
ALS
Rabin et al., 2009
Cryptic splicing-ASO approach to restore STATHMIN-2
Axonal degeneration and impaired repair
Healthy motor neuron
ALS motor neuron
Rescue of axonal stability and repair

- QurAlis QRL-201 is a therapeutic ASO that restores *STMN2* mis-splicing due to TDP-43 pathology
- We are developing biomarkers that can be used to identify patients with TDP-43 pathology and measure STMN2 target engagement

Stathmin-2 is a downstream target of TDP-43 function

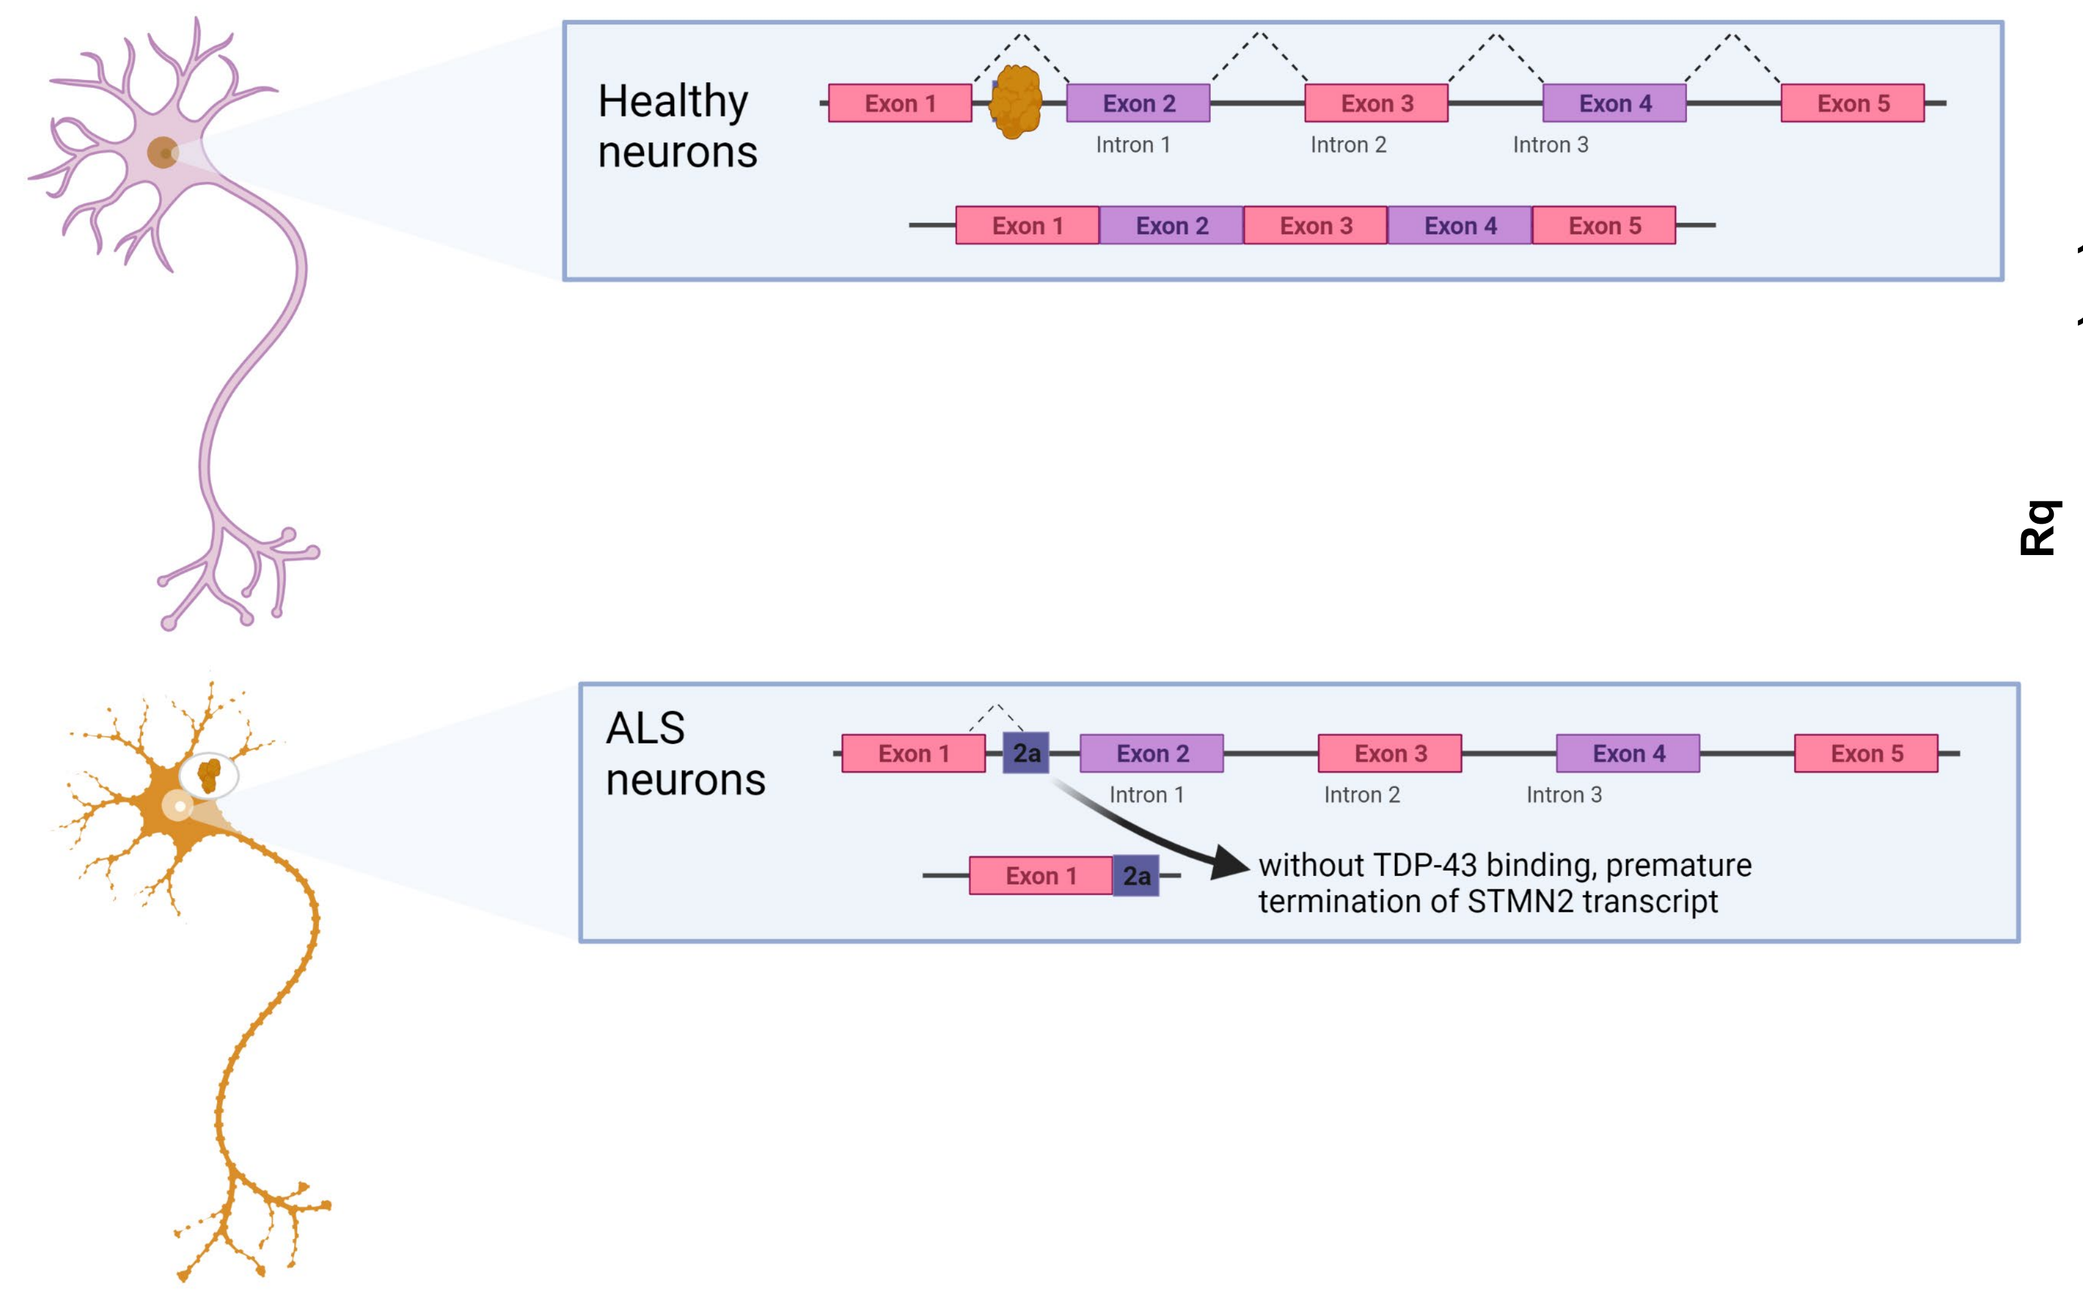
Healthy neurons
Exon 1
Exon 2
Exon 3
Exon 4
Exon 5
Intron 1
Intron 2
Intron 3
ALS neurons
2a
without TDP-43 binding, premature termination of STMN2 transcript

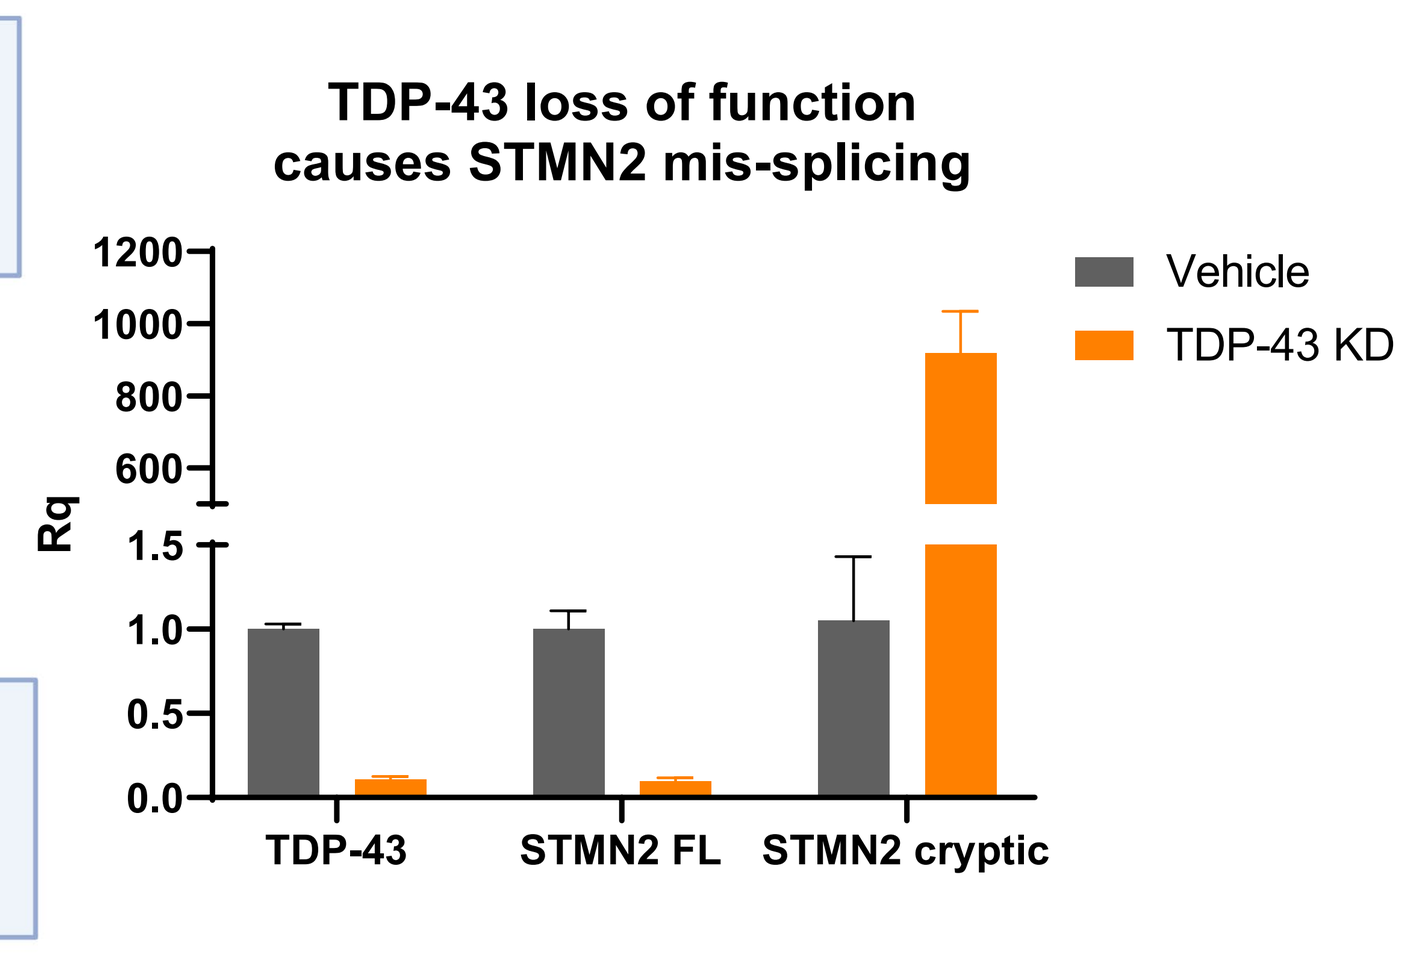
TDP-43 loss of function causes STMN2 mis-splicing
Rq
Vehicle
TDP-43 KD
TDP-43
STMN2 FL
STMN2 cryptic

Development of a Human Biofluid STMN2 RNA Detection Assay

qPCR and ddPCR assay development

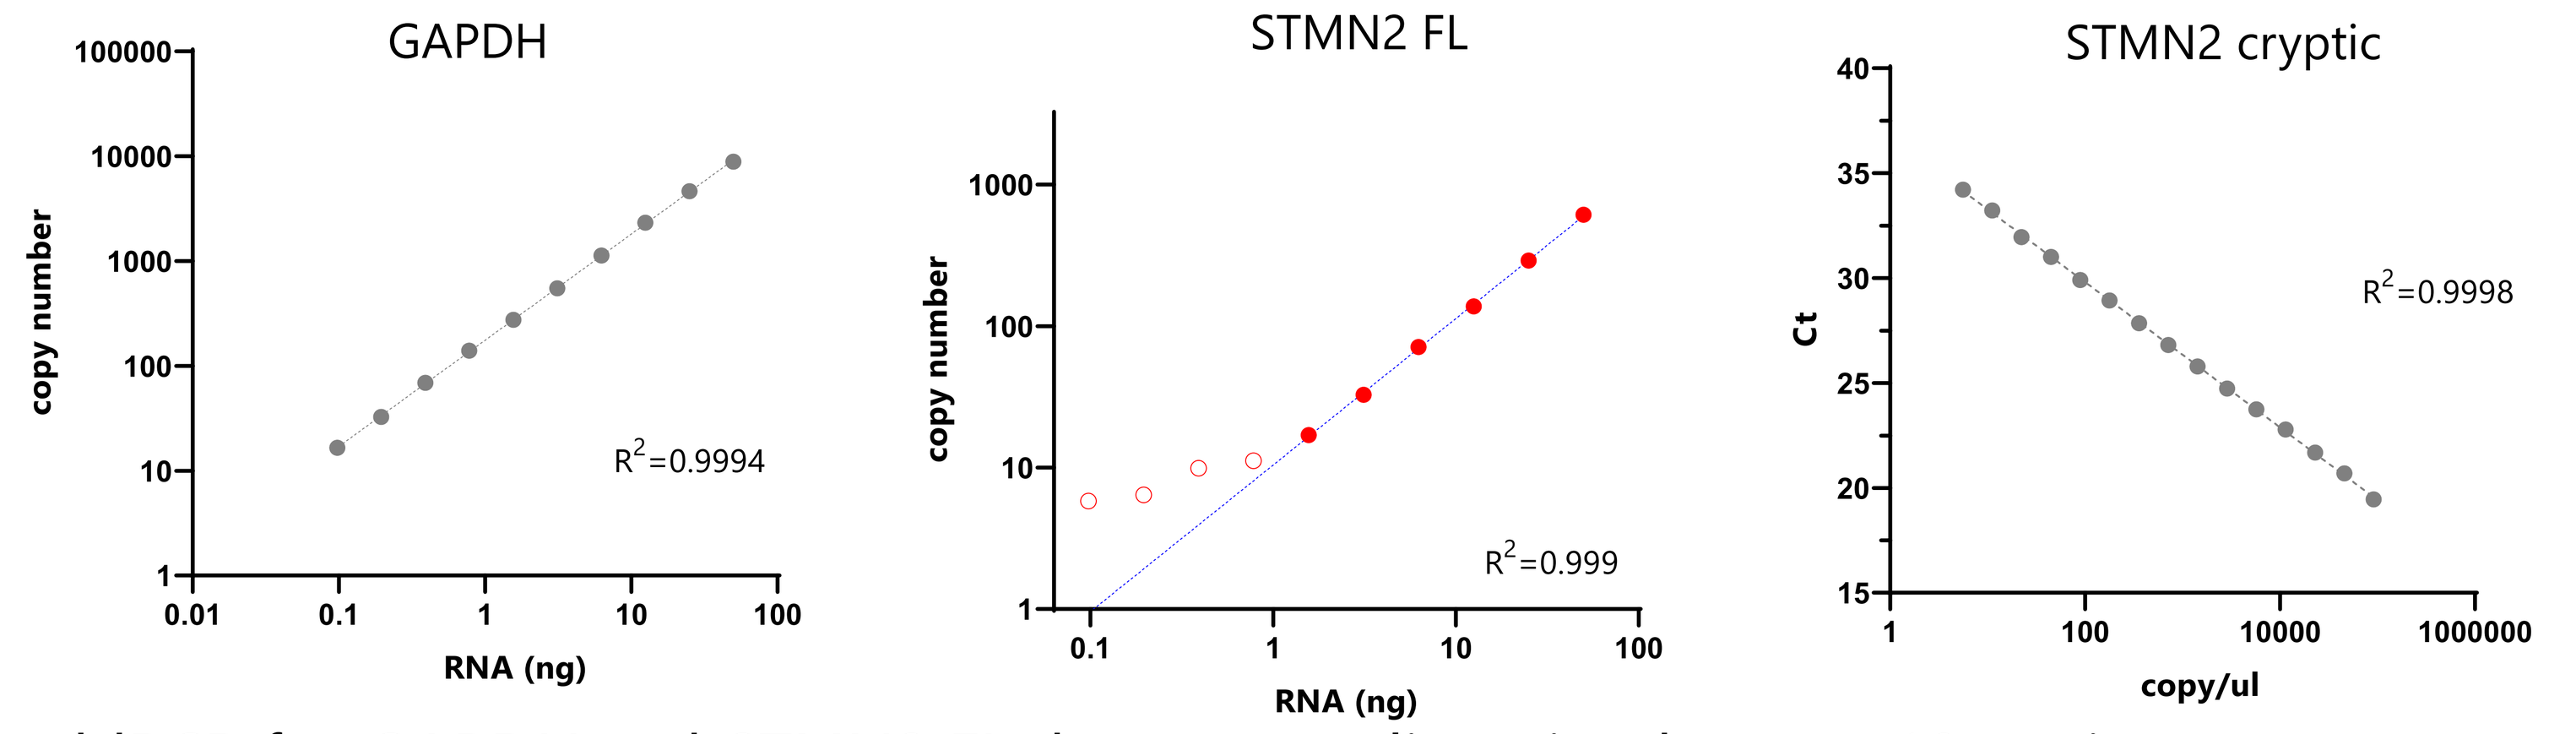
GAPDH
copy number
RNA (ng)
R^2=0.9994
STMN2 FL
R^2=0.999
STMN2 cryptic
Ct
copy/ul
R^2=0.9998

- ddPCR for *GAPDH* and *STMN2 FL* shows assay linearity down to 10 copies
- *STMN2* cryptic assay was evaluated using G-block DNA and qPCR testing shows assay linearity

Expression of *STMN2* RNA in hiPSC-derived conditioned media

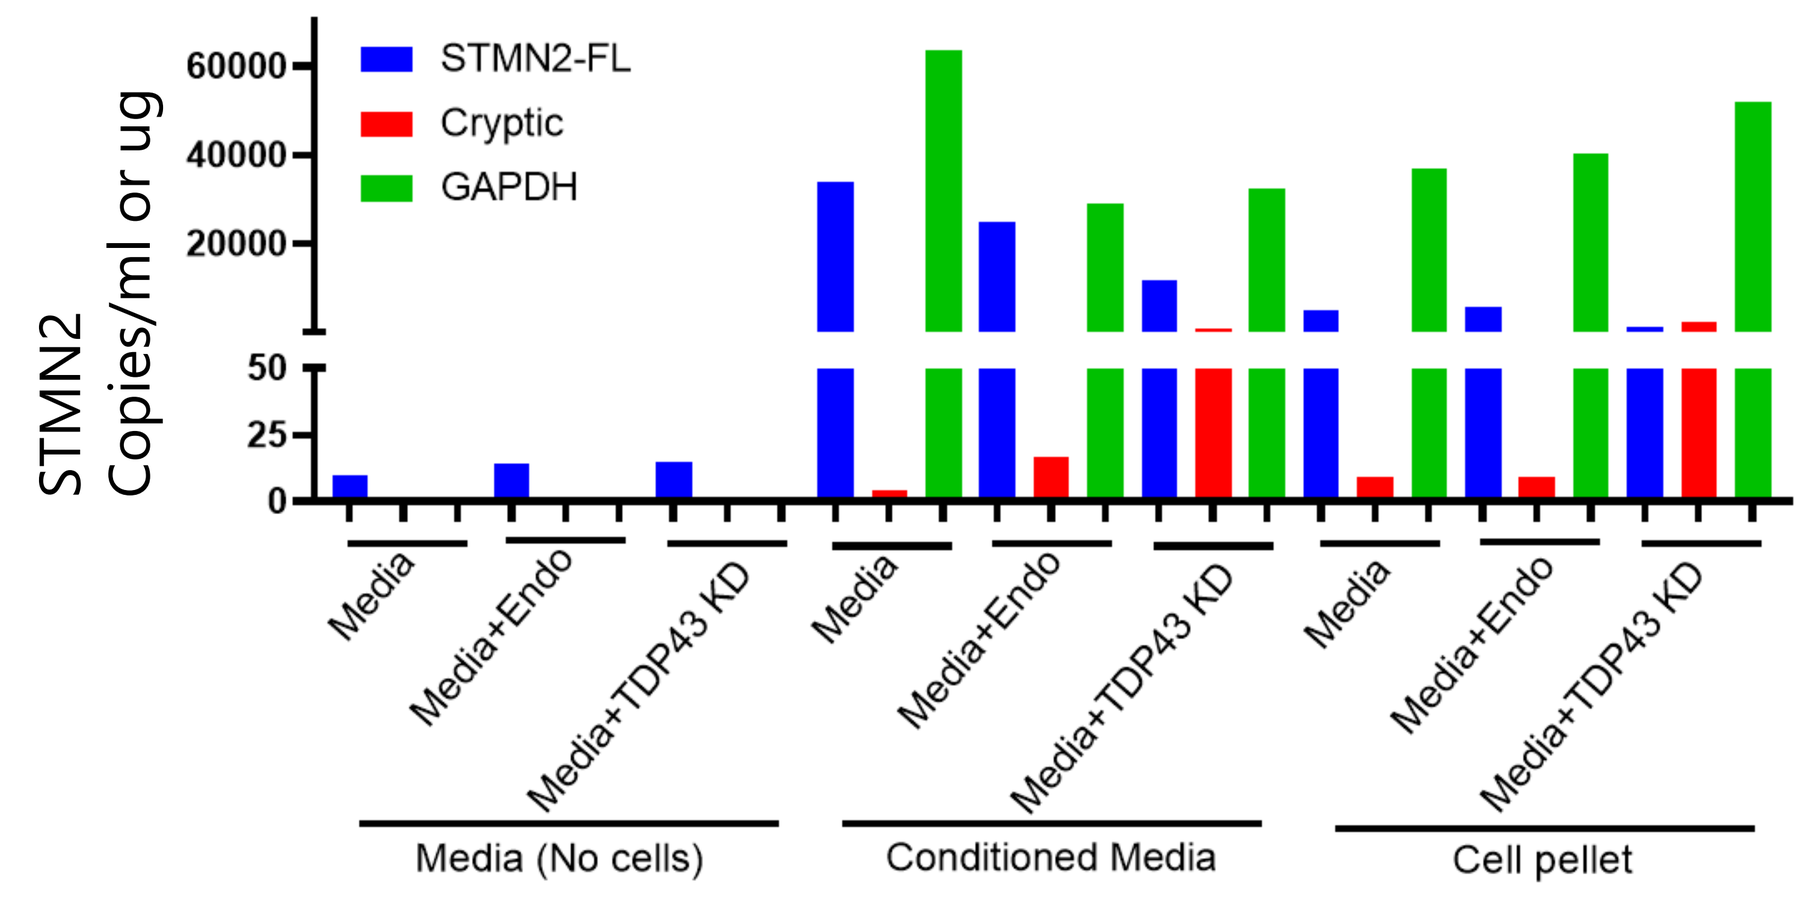
STMN2 Copies/ml or ug
STMN2-FL
Cryptic
GAPDH
Media
Media+Endo
Media+TDP43 KD
Media (No cells)
Conditioned Media
Cell pellet

- Quantitation of *STMN2 FL* and cryptic exon *STMN2* mRNA in exosomes isolated either from conditioned media or cell pellets of hiPSC-derived motor neurons treated with/out TDP-43 gapmer ASO to induce TDP-43 loss of function and *STMN2* mis-splicing
- Both *STMN2 FL* and *STMN2* cryptic exon mRNA is processed into motor neuronal exosomes
- Exosome *STMN2 FL* and cryptic levels represent the cellular biology and show *STMN2 FL* decrease and *STMN2* cryptic increase upon mis-splicing induction

STMN2 RNA quantification in human cerebrospinal fluid (CSF) and plasma

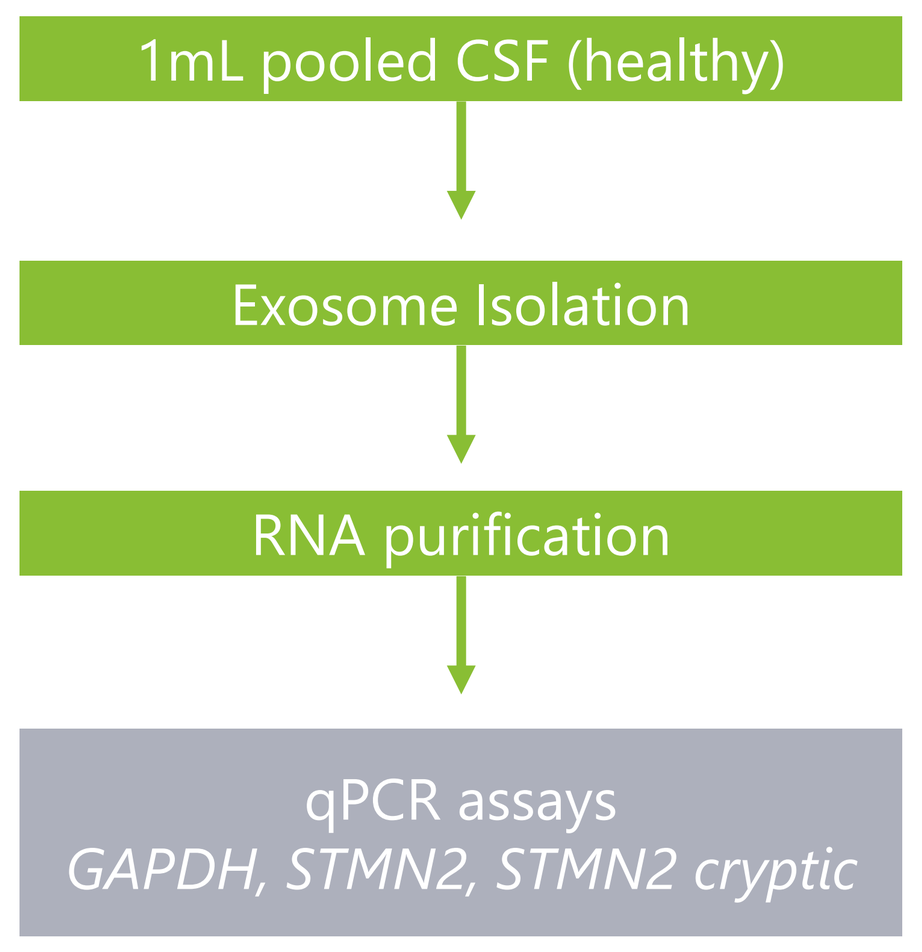
1mL pooled CSF (healthy)
Exosome Isolation
RNA purification
qPCR assays
GAPDH, STMN2, STMN2 cryptic

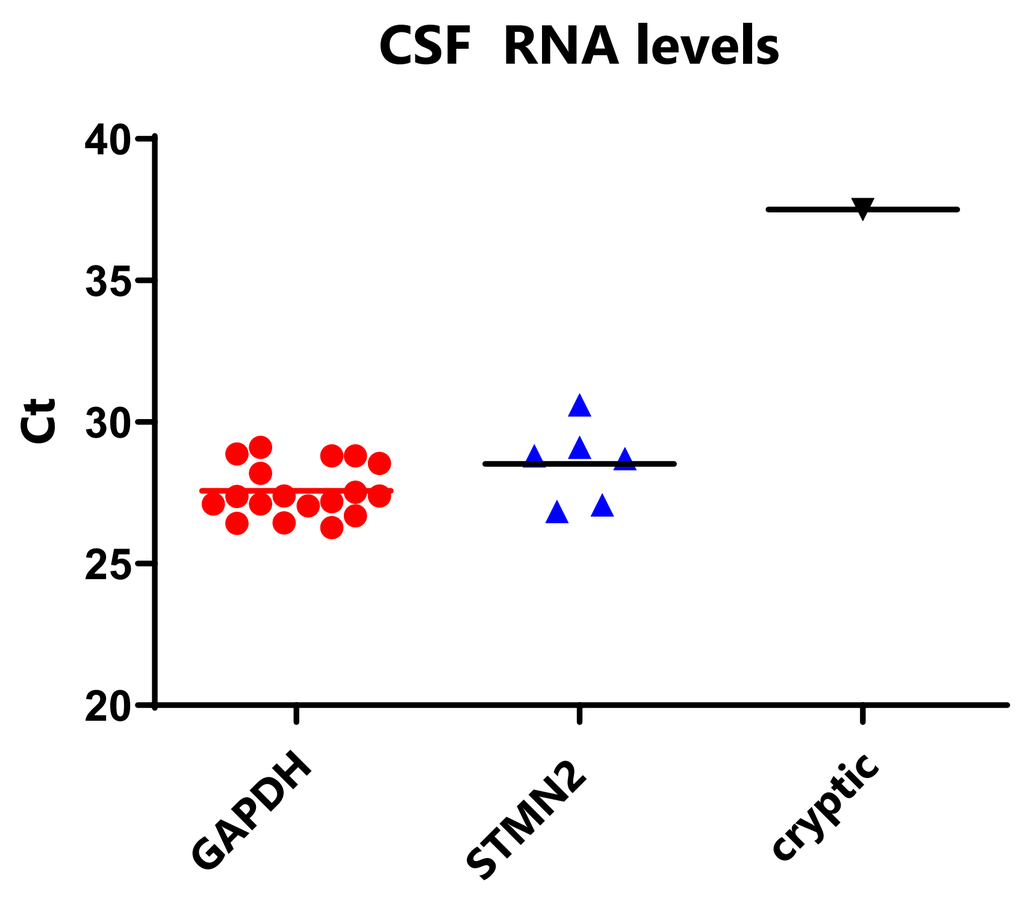
CSF RNA levels
Ct
GAPDH
STMN2
cryptic

- Healthy CSF was assayed for the presence of *STMN2* RNA
- *STMN2* RNA was detected at high quantity in CSF exosomes, about the same as the reference *GAPDH*
- Cryptic exon containing transcripts were mainly not detected, as expected in healthy samples

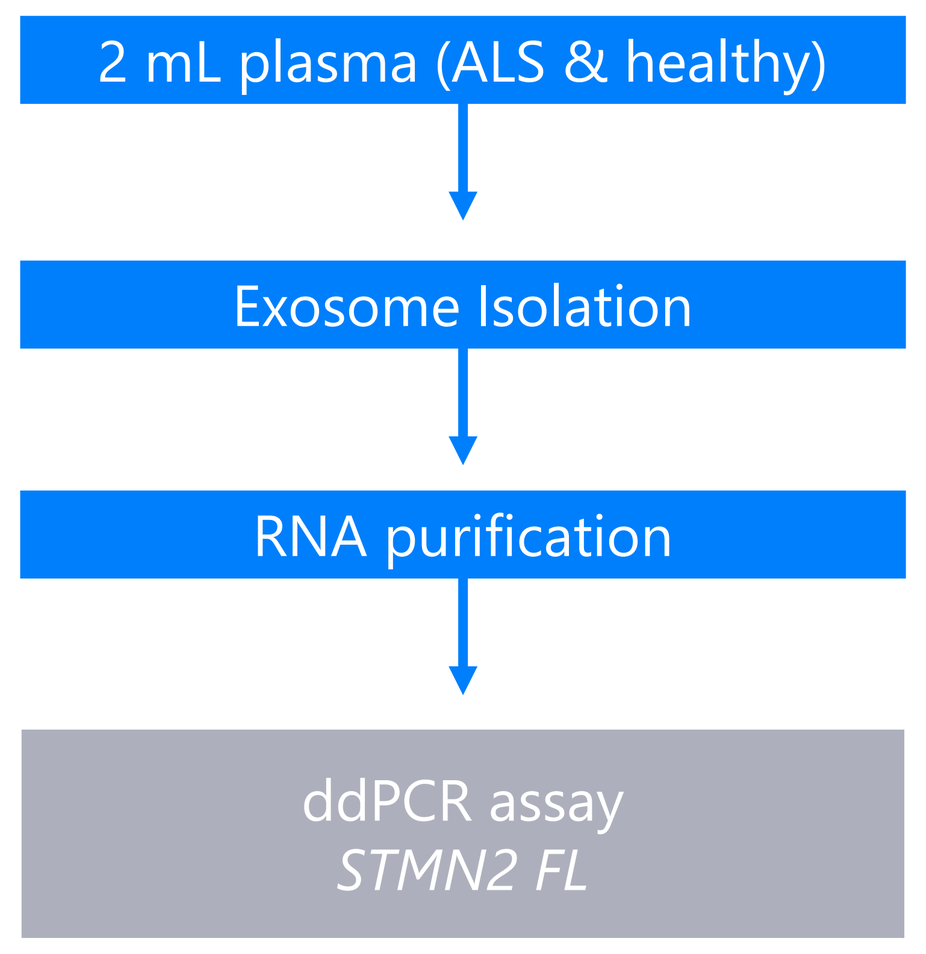
2 mL plasma (ALS & healthy)
Exosome Isolation
RNA purification
ddPCR assay
STMN2 FL

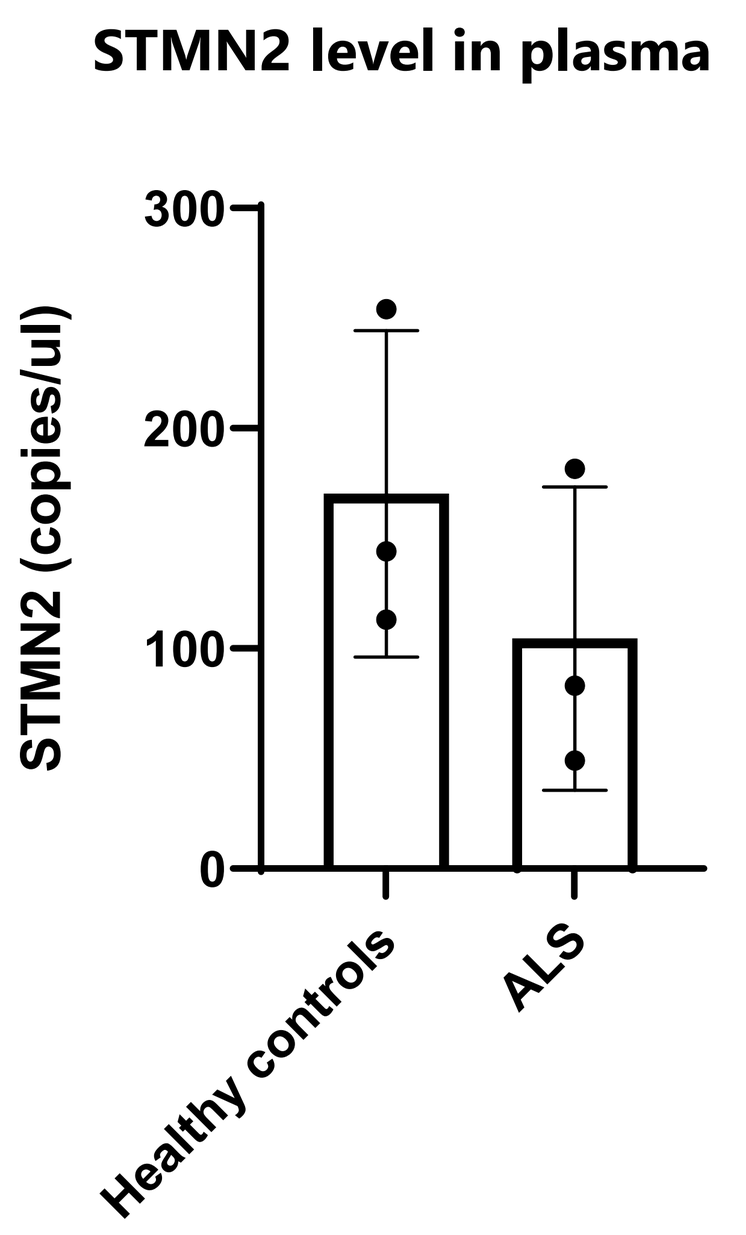
STMN2 level in plasma
STMN2 (copies/ul)
Healthy controls
ALS

n=3, Mean±SD
unpaired t-test
NS, p=0.32

- ALS and healthy plasma was assayed for the presence of *STMN2* RNA
- *STMN2* RNA was detected in plasma exosomes
- Trend toward decreased *STMN2 FL* RNA level in ALS plasma exosomes

Development of a human biofluid STMN2 Protein Detection Assay

SIMOA assay development

Calibration Curve			
[stathmin-2] pg/mL	Average AEB	%CV	S/B
0.0	0.005	22%	
0.4	0.012	2%	2
1.5	0.034	8%	6
5.9	0.120	10%	22
23.4	0.450	13%	82
93.8	2.282	13%	417
375.0	8.023	0%	1466
1500.0	22.500	2%	4111

LOD: 0.087 pg/ml
LLOQ: 0.33 pg/ml

- STMN2 capture and detection antibody pairs were tested and the 2 with lowest S/B and dilution linearity were selected
- The final assay format shows low background levels (AEB<0.01) and sub-pg/mL LLOQ

STMN2 SIMOA assay detects disease relevant changes in iPSC-derived motor neuron lysates

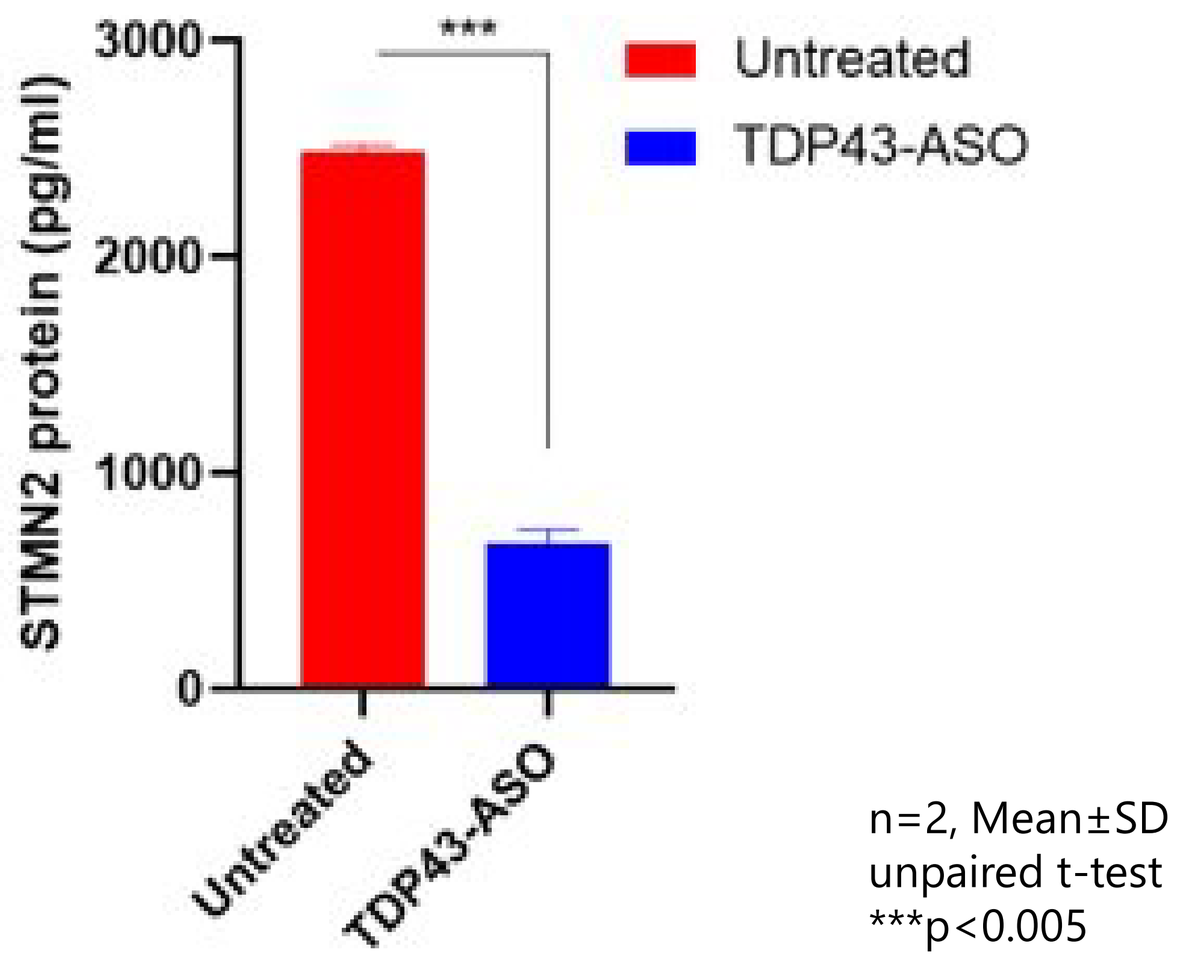
STMN2 protein (pg/ml)

Untreated
TDP43-ASO

n=2, Mean±SD
unpaired t-test
***p<0.005

- hiPSC-derived motor neurons were treated with/out TDP-43 gapmer ASO
- TDP-43 KD results in STMN2 mis-splicing and loss of STMN2 protein levels
- STMN2 SIMOA assay detects changes in human STMN2 levels due to TDP-43 loss of function

STMN2 protein quantification in human plasma

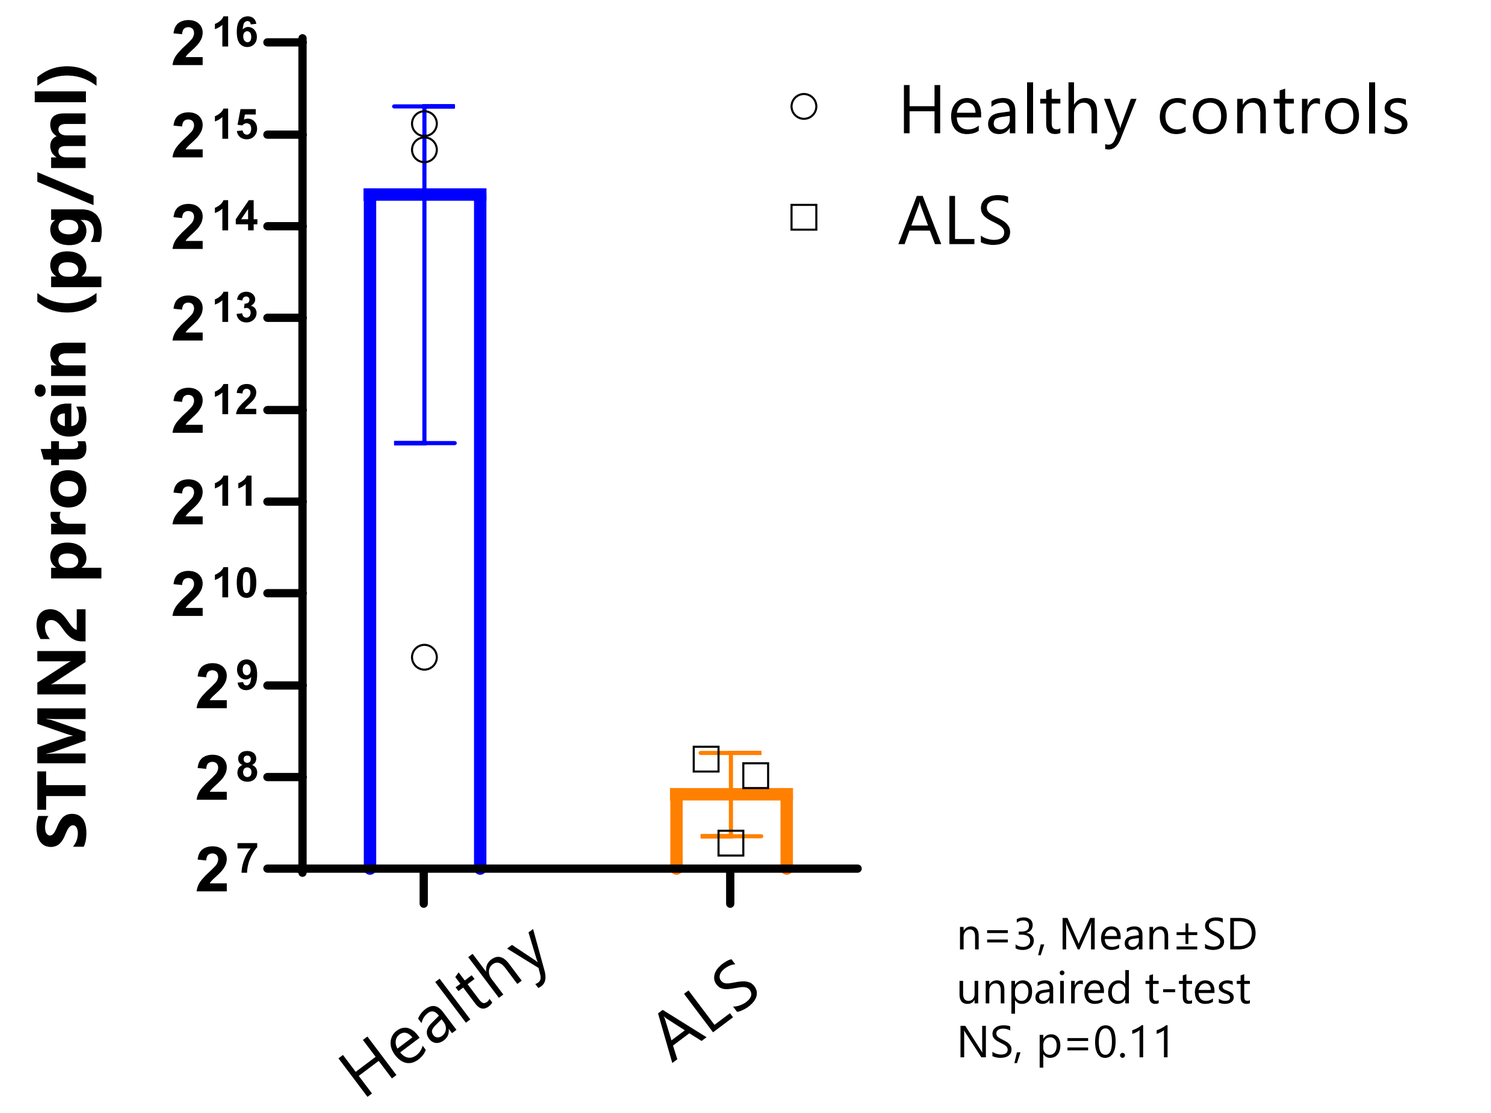
STMN2 protein (pg/ml)
Healthy controls
ALS
Healthy
ALS

n=3, Mean±SD
unpaired t-test
NS, p=0.11

- Healthy and ALS plasma was tested in the STMN2 SIMOA assay (n=3 individuals each)
- Healthy control and ALS samples within the dynamic range of the assay
- ALS STMN2 levels are 50% reduced compared to the lowest healthy sample and 98% reduced compared by mean values

Conclusions

- We have developed both RNA and protein quantitation methods to detect STMN2 in human biofluids
- STMN2 biomarker assay allows the characterization of TDP-43 pathology in living ALS patients
- Next steps include:
 - Assay qualification and validation for use as an exploratory Biomarker
 - Characterization of larger patient cohorts from annotated clinical libraries
 - Increased sensitivity in RNA assays for optimization of cryptic exon quantitation in CSF and plasma

Acknowledgements

Work supported in part by: Target ALS Industry Led Consortium Grant

References

1. Klim and Williams et al *Nat. Neurosci* 22, 167-179 (2019).
2. Rabin S. J. et al *Hum. Mol. Genet.* 19, 313-328 (2009).